Feasibility of establishing and drug screening patient-derived rectal organoid models from pretreatment rectal cancer biopsies

Scarlett Acklin-Wehnert*[1], Divya Dayanidhi*[2], Brian G. Czito[1], Manisha Palta[1], Christopher Willett[1], Christine Elissa Eyler[1], Christopher Mantyh[3], John Migaly[3], Julie Thacker[3], Billy Lan[3], David S. Hsu[4]
Department of Radiation Oncology[1] and Chemistry[2], Division of Surgical Oncology[3] and Medical Oncology[4], Duke University

Background:
Response to neoadjuvant chemotherapy and radiation therapy in the treatment of locally advanced rectal cancer is heterogenous and prognostic of clinical outcomes, necessitating the need for predictive biomarkers to guide personalized treatment recommendations. Sensitivity to a given chemotherapy in patient-derived organoids predicts patient response to that chemotherapy, establishing it as a promising model for efforts to ascertain predictive biomarkers and personalize treatment decisions. This study assessed the feasibility of obtaining patient-derived rectal organoids from standard of care pre-treatment proctoscopy biopsies.

Methods:
In this clinical trial (NCT04371198), biopsies were obtained from patients with stage II rectal adenocarcinoma prior to receipt of neoadjuvant therapy. Tissue samples were mechanically and enzymatically dissociated to obtain a single cell suspension. Cells were then mixed with matrigel at a ratio of 2,000 cells:5 µL Matrigel in a 50 µL dome and plated on a 24-well tissue culture plate with colorectal cancer organoid media at 37°C/5% CO_2. Established patient-derived organoids were then used to perform drug screens with clinically-applicable chemotherapeutics including oxaliplatin, irinotecan, and 5-FU, followed by high throughput drug screen using our recently published MicroOrganoSpheres platform using the NCI Approved Oncology Drugs Set VI library.

Results:
Of the 20 patients enrolled, 17 (85%) patient-derived organoids were created from pre-treatment specimens. 15 (88%) of these samples were successfully established as defined by the ability to passage organoids for at least two passages. All established samples were used to perform standard of care drug screens and high throughout drug screens, which demonstrated differences in drug sensitivities among the samples. Moreover, within two weeks of receiving the sample, four established quickly enough to complete drug screening with oxaliplatin, SN38, and 5-Fluorouracil.

Conclusions:
These results demonstrate the feasibility of establishing patient-derived rectal organoids from biopsy specimens obtained by proctoscopy, and reinforce the utility of patient-derived organoids as a tractable ex vivo platform to personalize rectal cancer treatment. Planned future directions include in vitro determination of radiation therapy sensitivity as well as systematic assessment of the correlation between individual patients and their organoid model.

Clinical Protocol and Methods

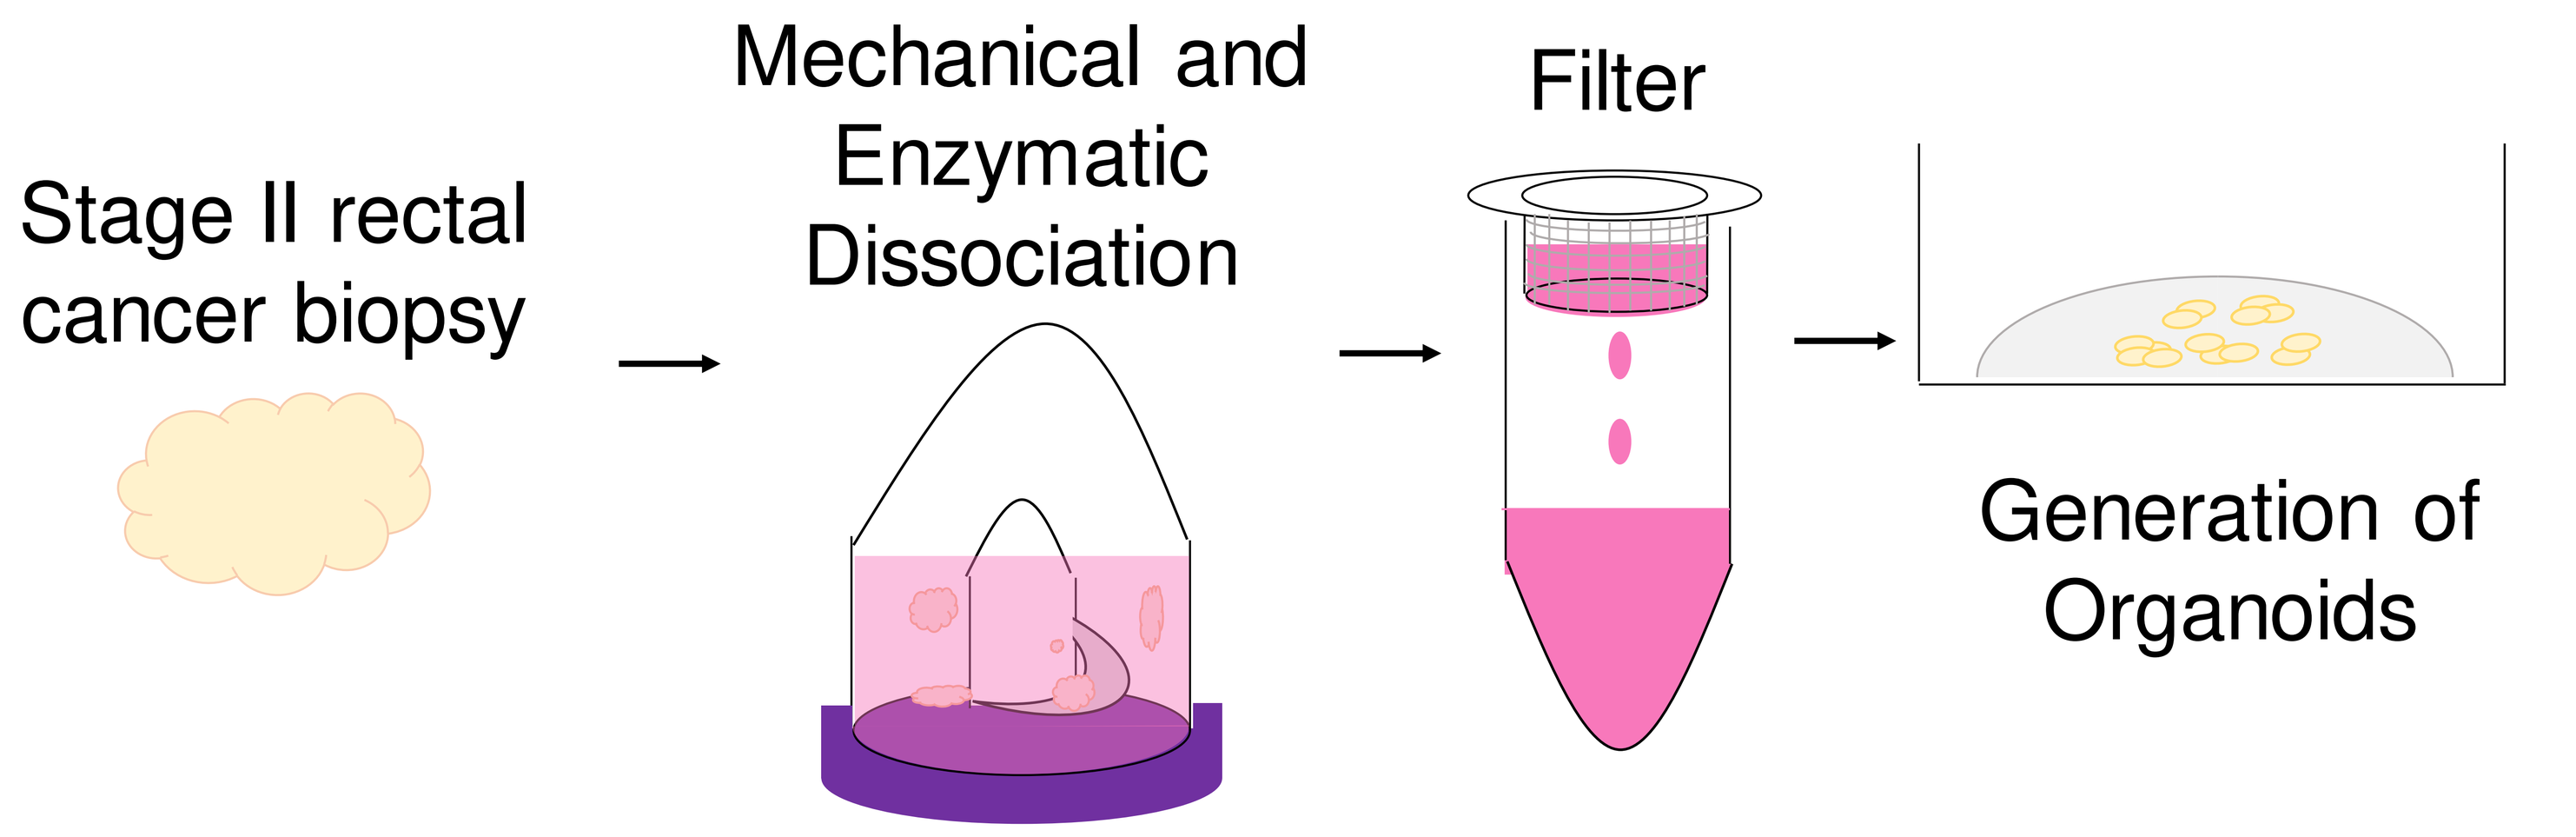
Stage II rectal cancer biopsy
Mechanical and Enzymatic Dissociation
Filter
Generation of Organoids

Goal: Establish patient derived organoids and perform drug screens within two weeks of obtaining rectal cancer biopsies

Rectal Cancer Organoids Established from Biopsies

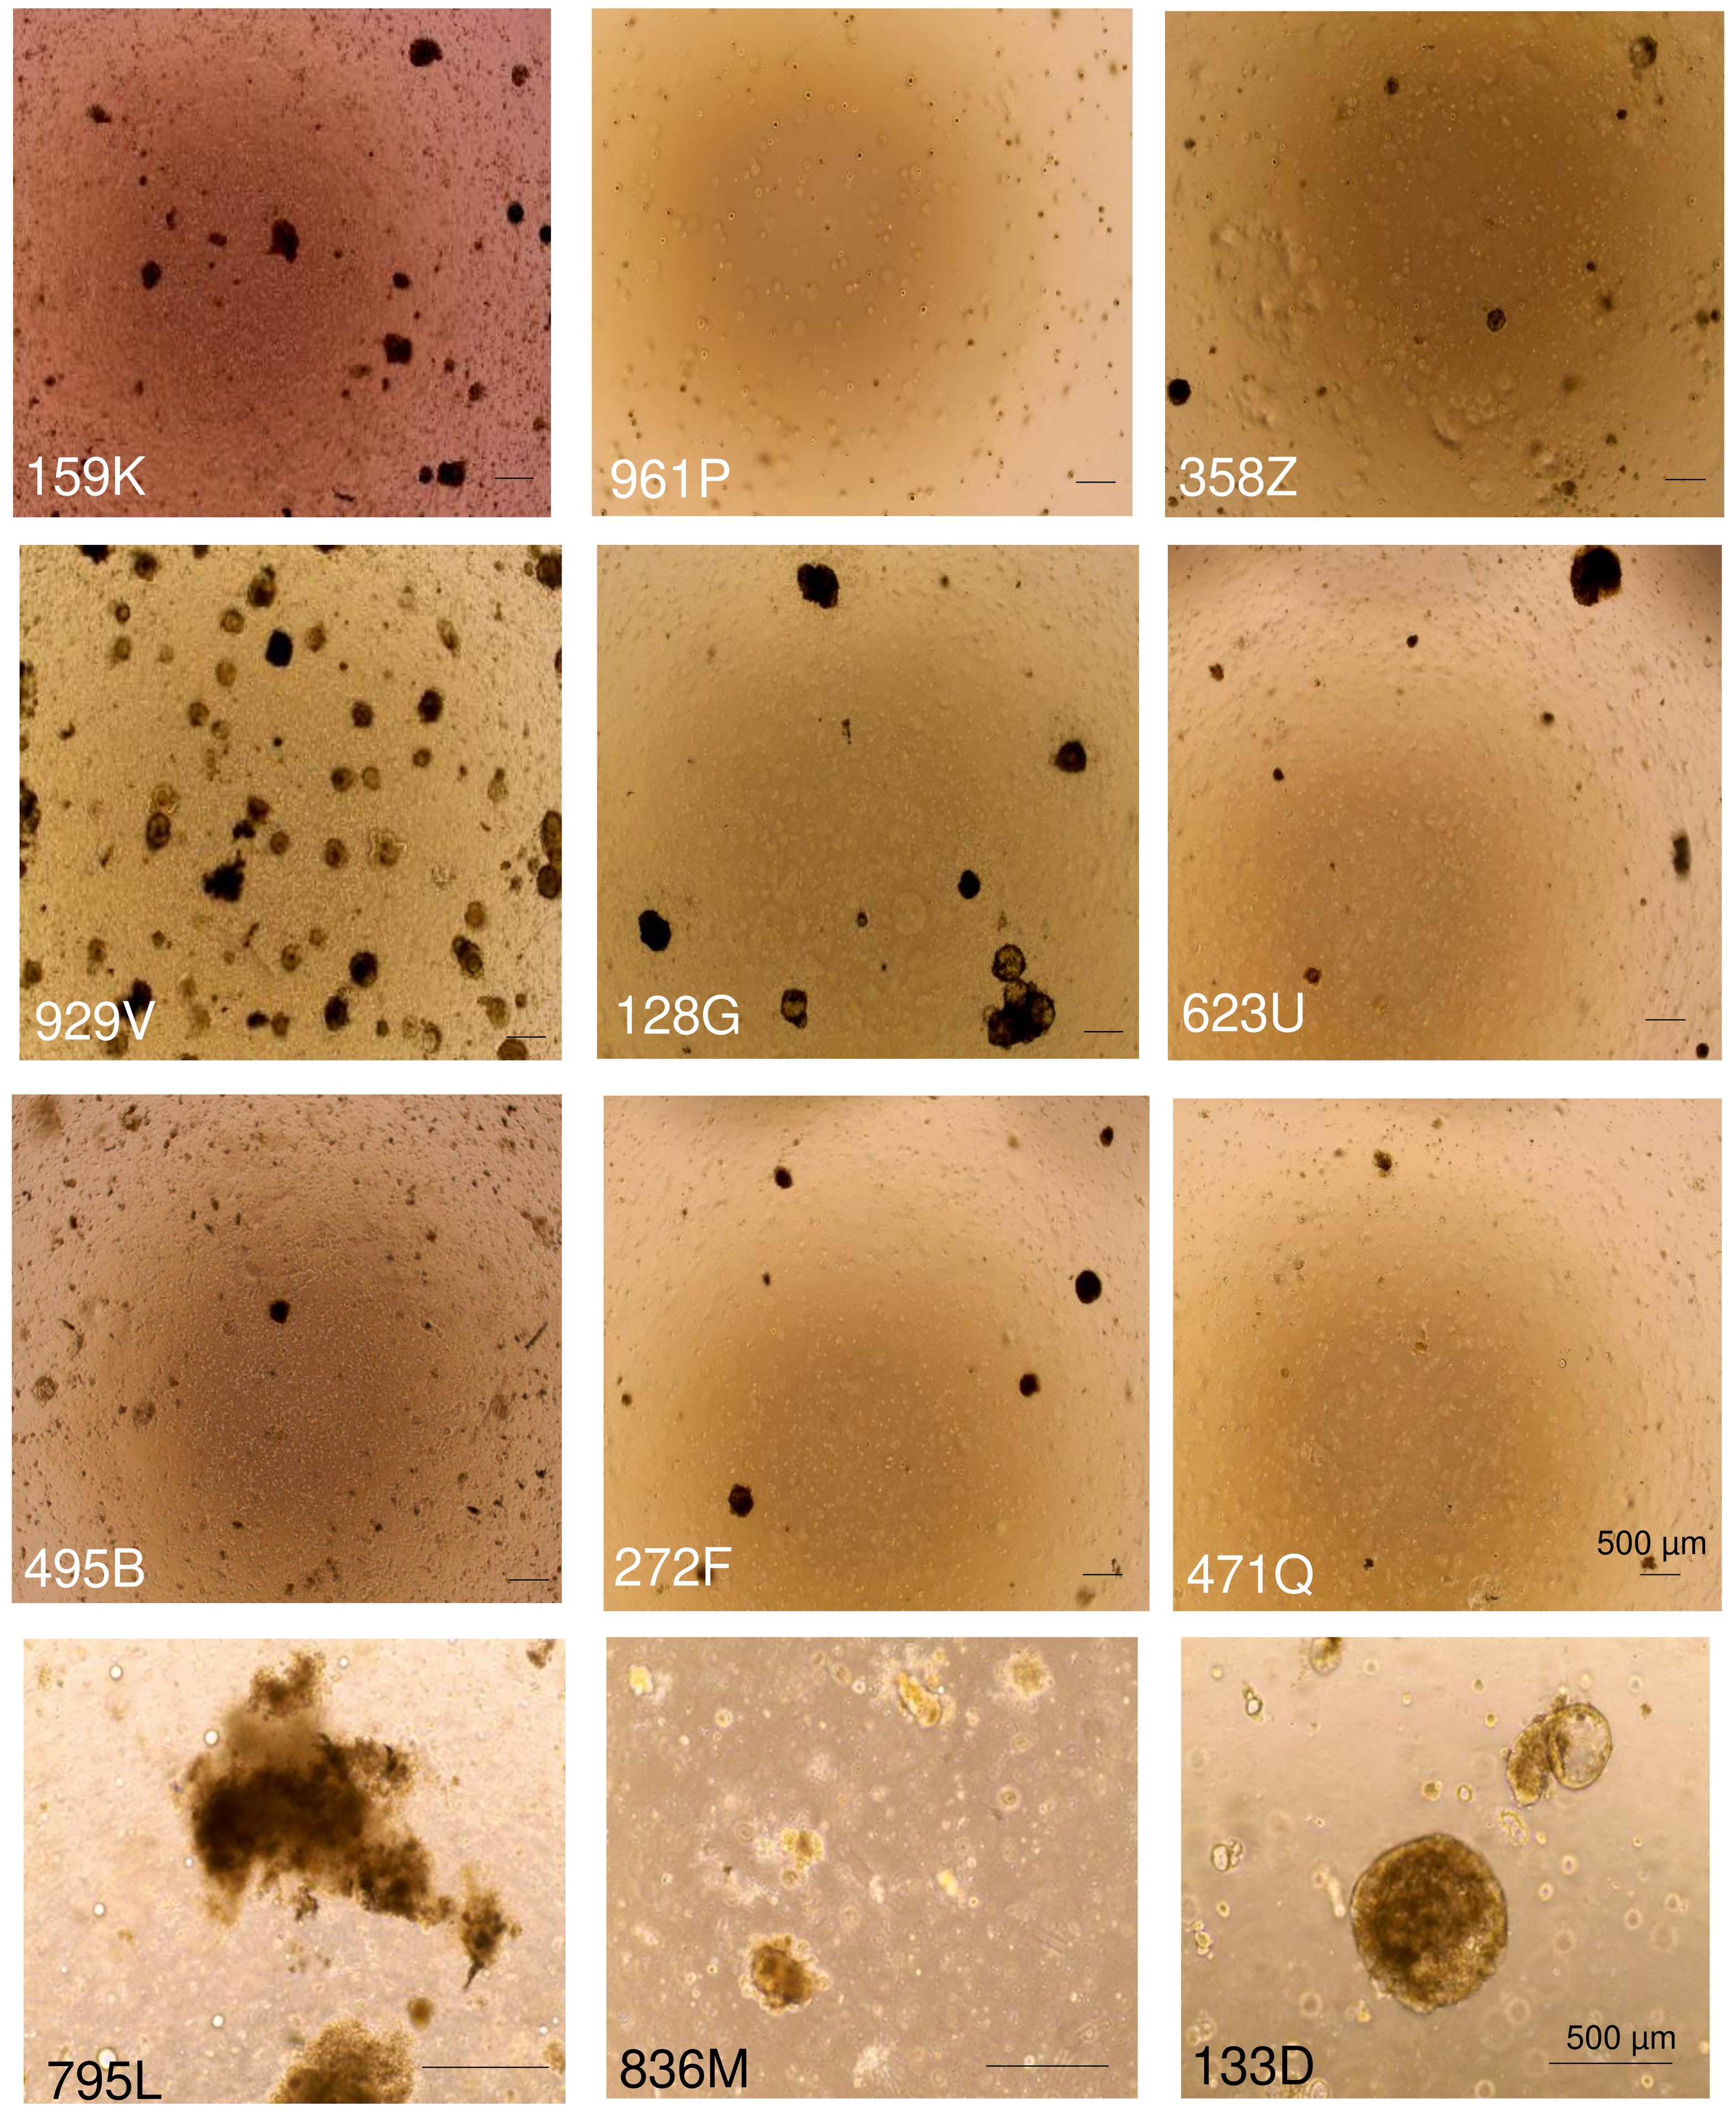
159K
961P
358Z
929V
128G
623U
495B
272F
471Q
500 µm
795L
836M
133D
500 µm

Figure 1: Establishment of rectal cancer organoids from biopsy samples. Representative images of rectal cancer organoids growing in 50 µL Matrigel domes. Images taken at 4X (Top Three Row) or 10X (Bottom Row) on an EVOS microscope.

High Throughput Drug Screens on Rectal MOS

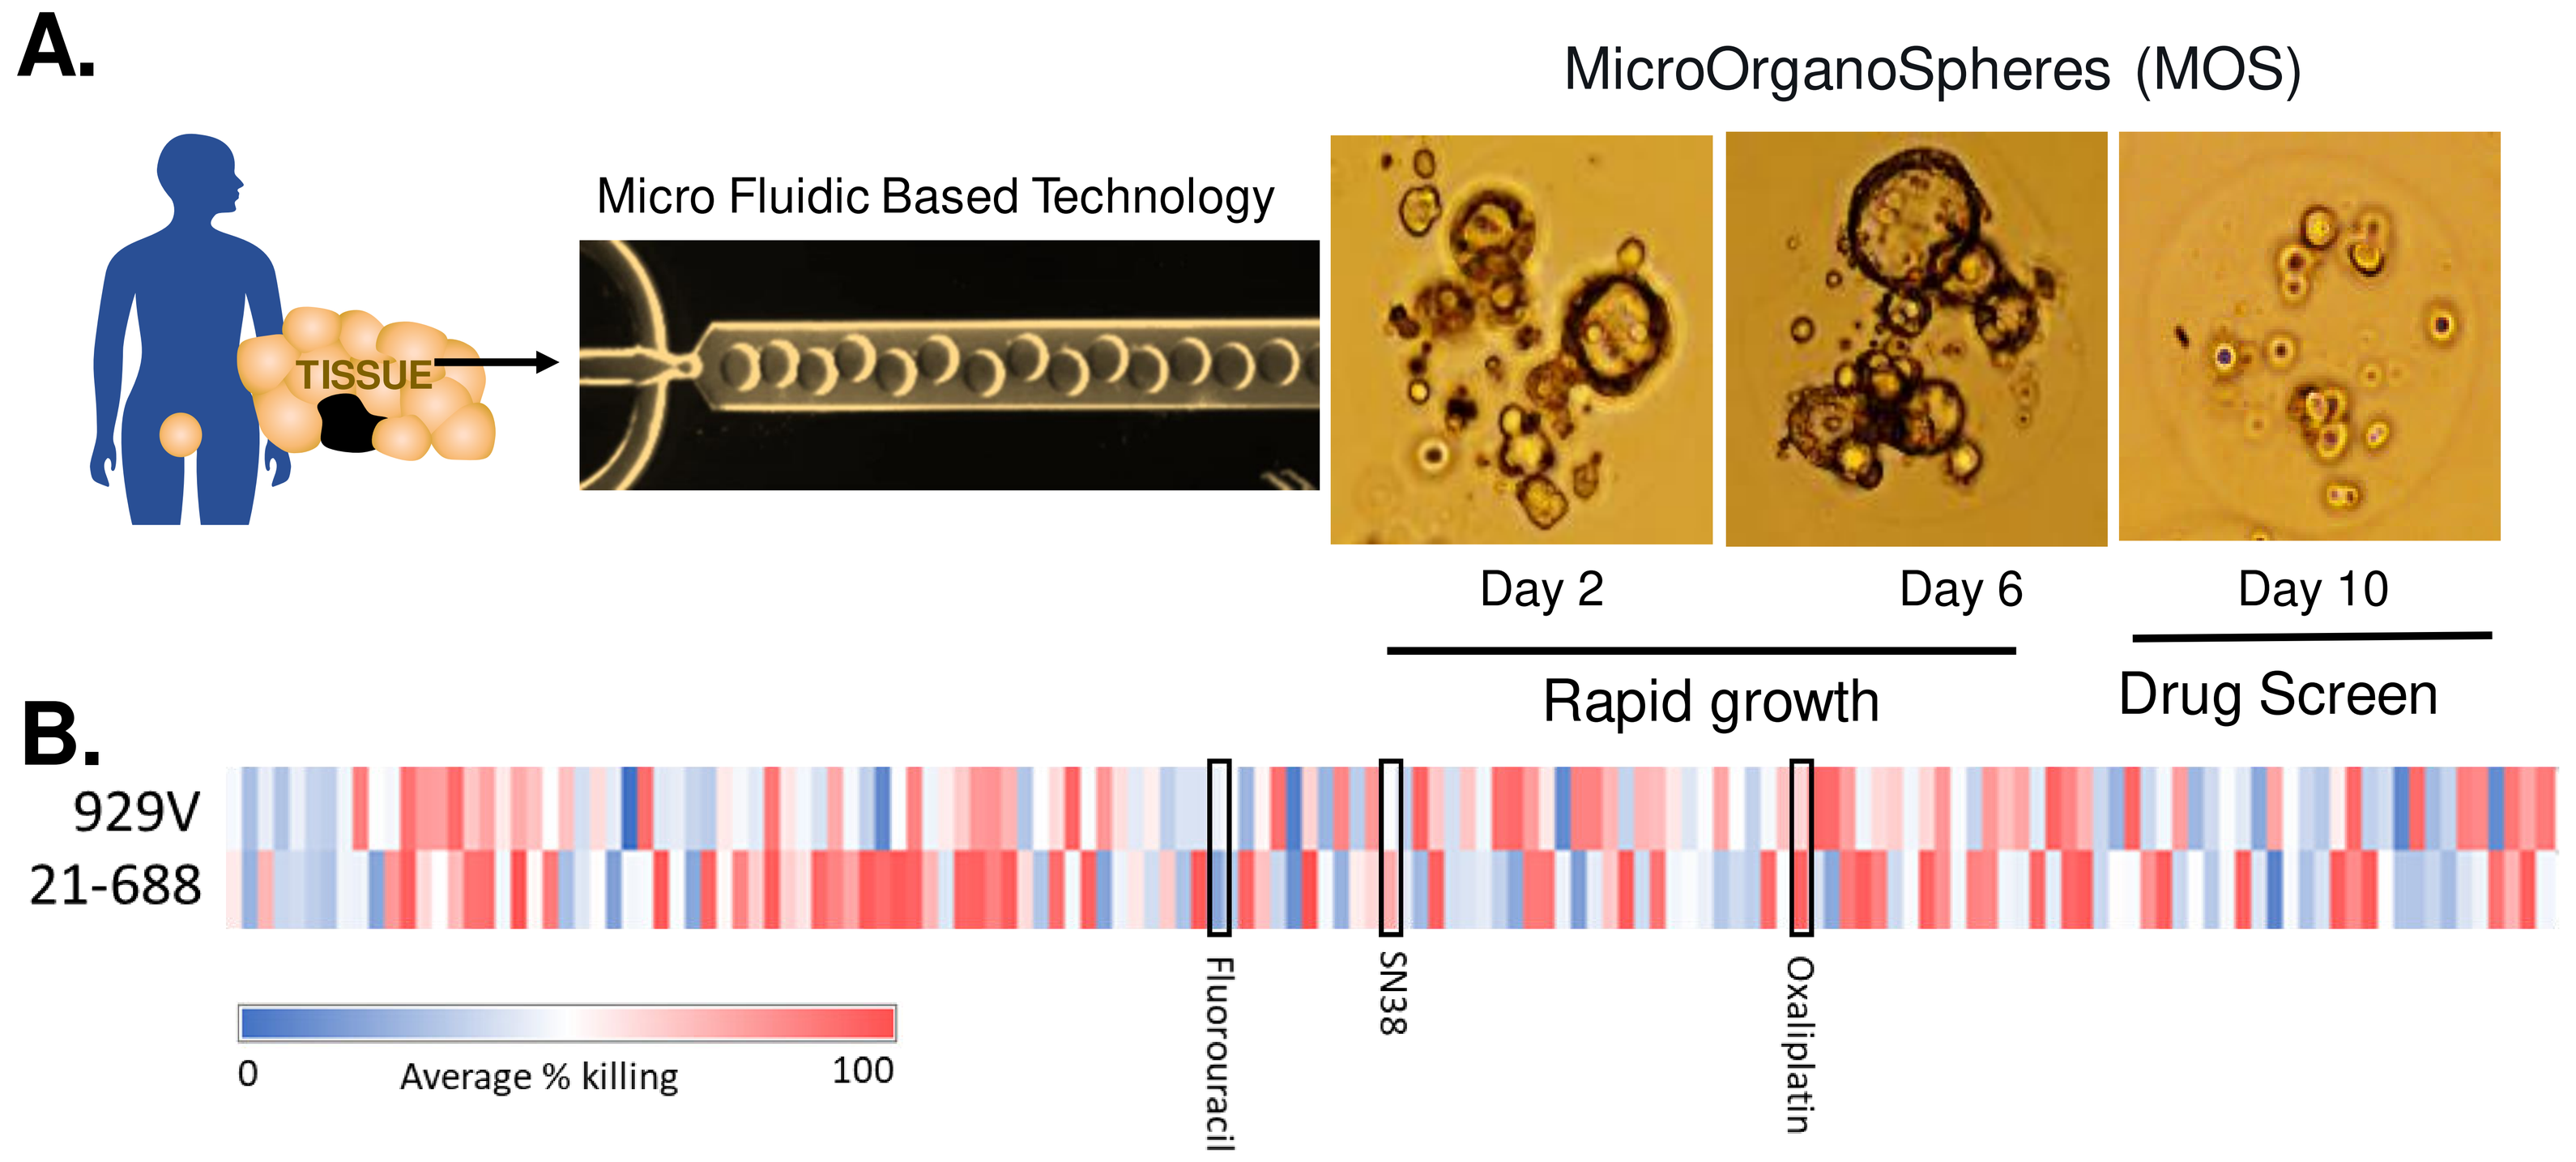
A.
MicroOrganoSpheres (MOS)
Micro Fluidic Based Technology
TISSUE
Day 2
Day 6
Day 10
Rapid growth
Drug Screen
B.
929V
21-688
0
Average % killing
100
Fluorouracil
SN38
Oxaliplatin

Figure 3: MicroOrganoSpheres (MOS) facilitates Drug Screen. A) Using microfluidic based technology, MOS can be generated in 10d for drug screens. B) High throughput drug screen of rectal MicroOrganSphere (MOS) across 147 FDA approved cancer drugs reveals differing drug sensitivity

Single-Agent Drug Screens of Rectal Organoids

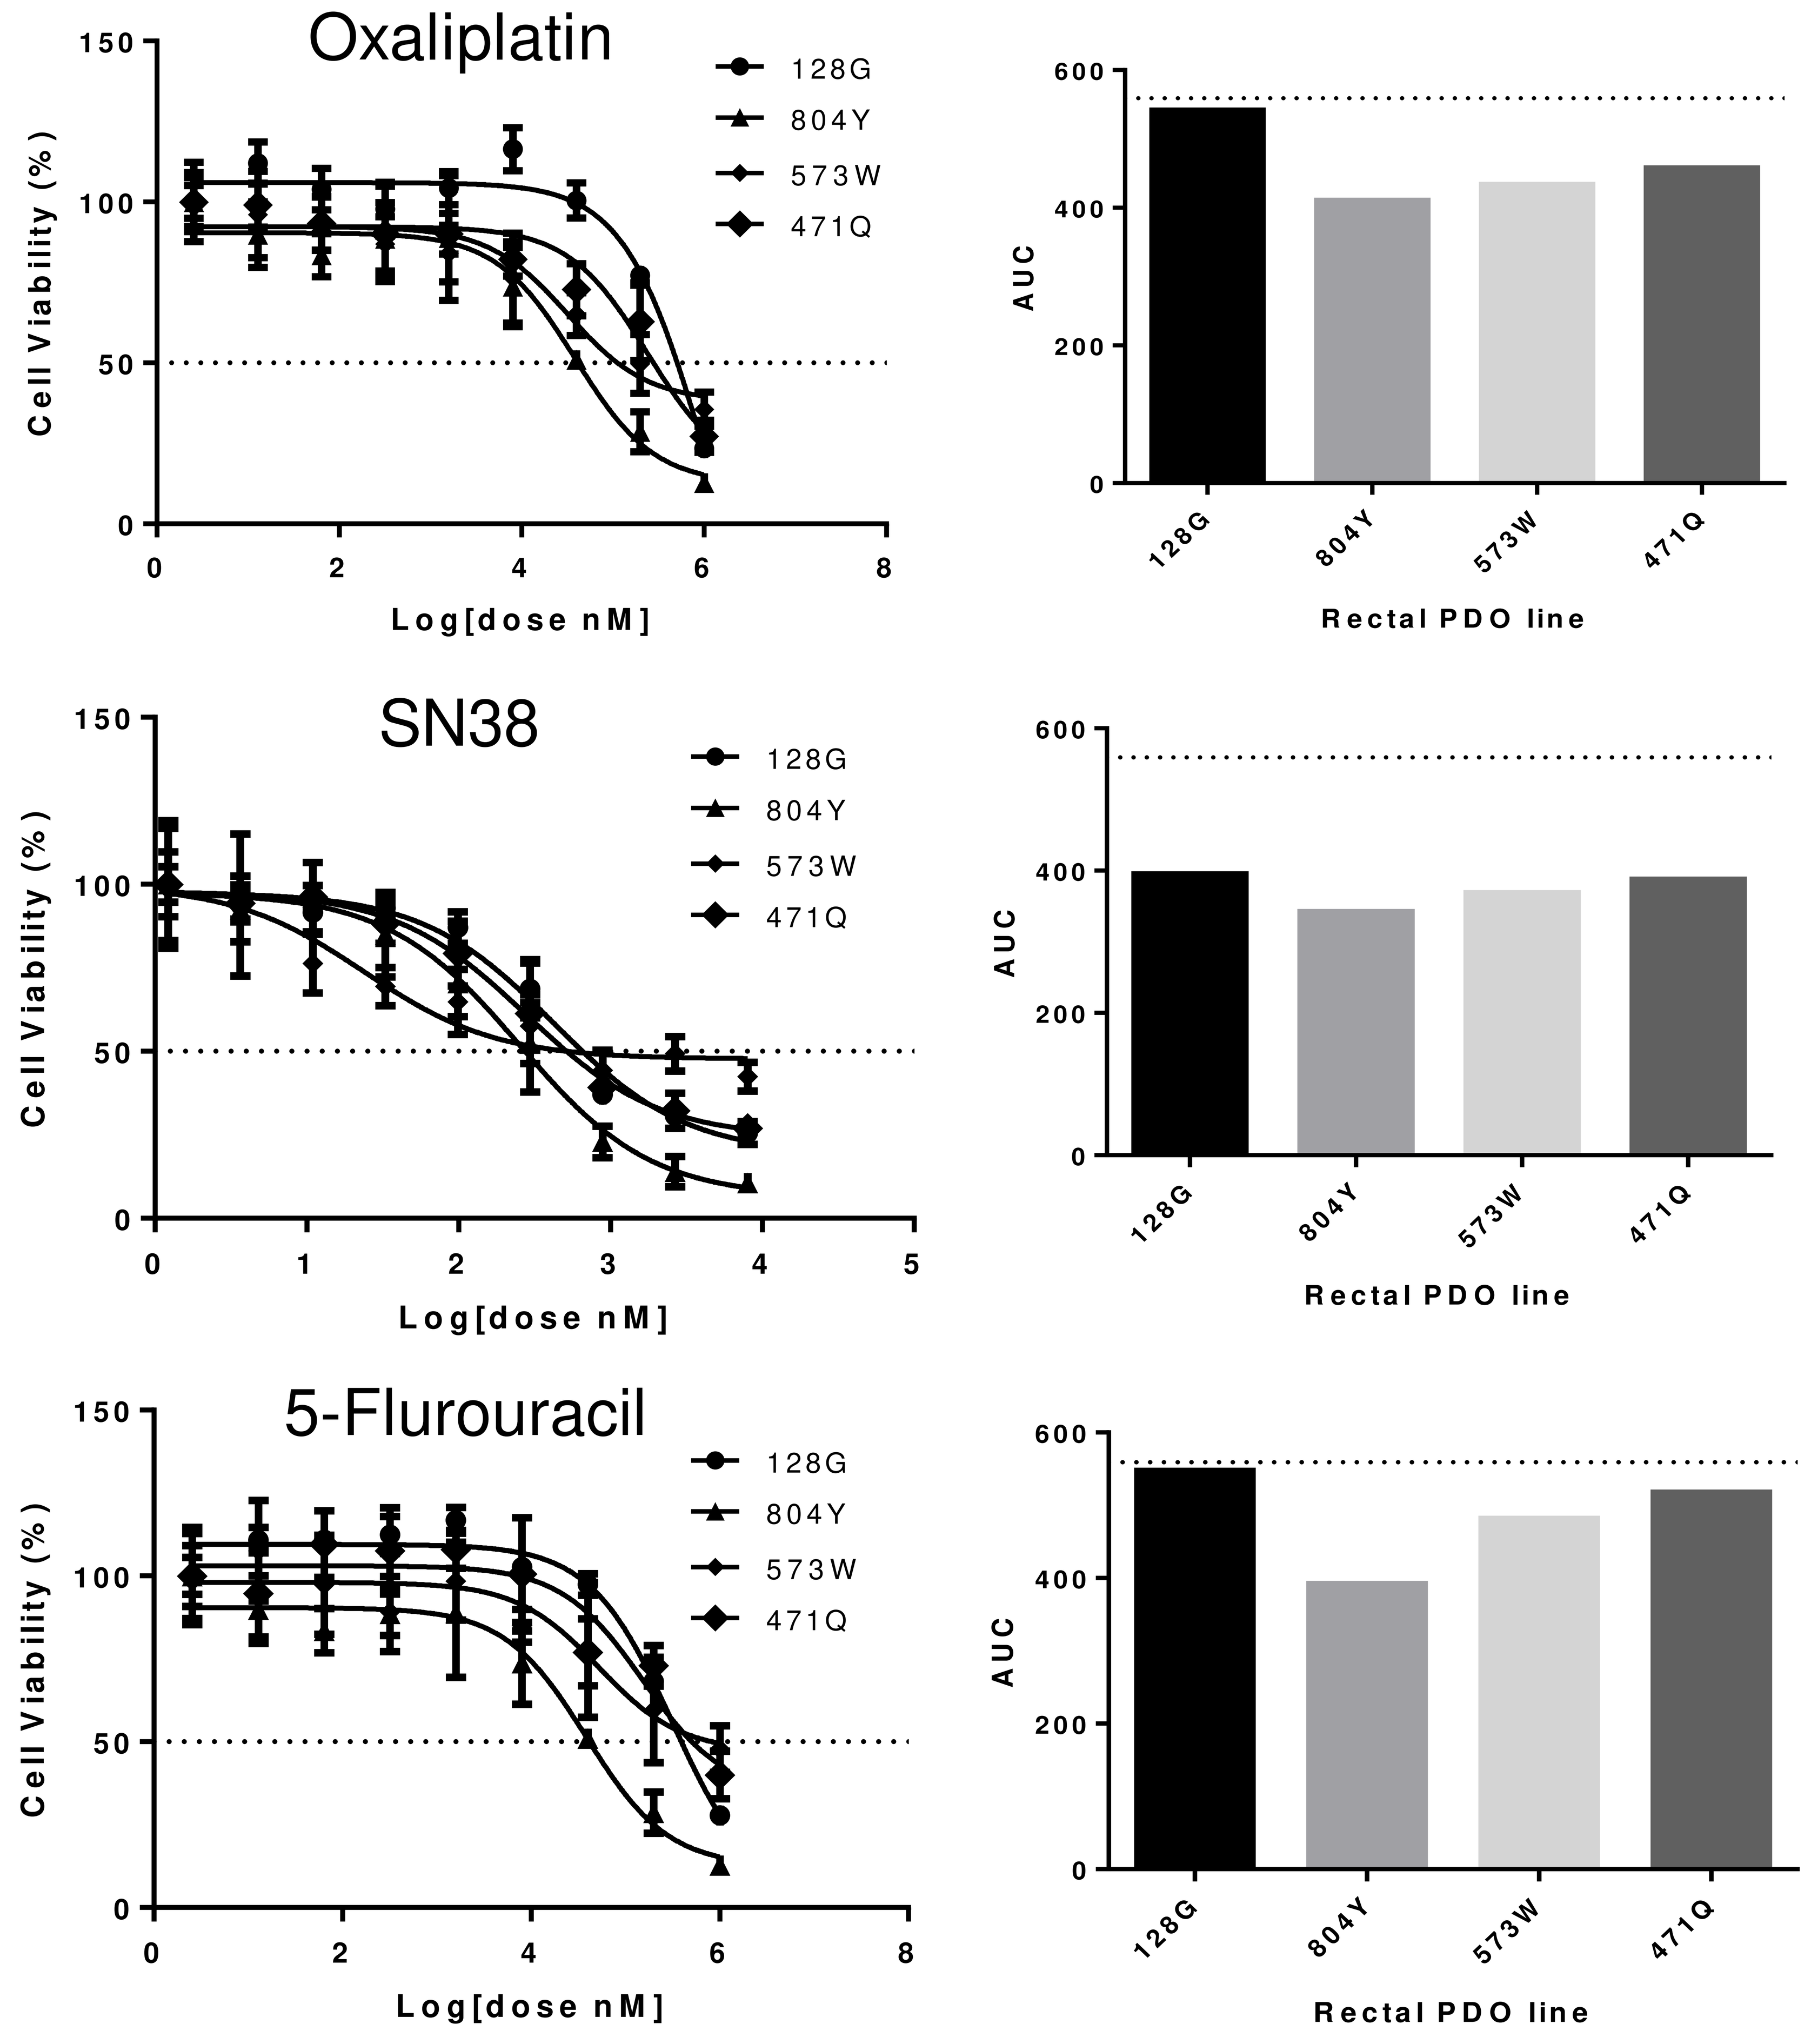
Oxaliplatin
SN38
5-Flurouracil
128G
804Y
573W
471Q
Cell Viability (%)
Log[dose nM]
AUC
Rectal PDO line

Figure 2: Single agent drug screens for oxaliplatin, SN38, and 5-Fluorouracil within two weeks of receiving biopsy samples. Drug screens were completed with nine concentrations per drug and five replicates per concentration. Organoids were incubated with drug for three days before adding Cell Titer Glo as a measure of cell viability.

Future Directions

- Continue high-throughput drug screens on established rectal organoids using the NIH147 panel of FDA-approved drugs
- *in vitro* and *in vivo* determination of radiation therapy sensitivity
- Systematic assessment of the correlation between individual patients and their organoid model.

References

Ding S, Hsu C, Wang Z, et al., Patient Derived Micro-OrganoSpheres enable Clinica; Precision Oncology. Cell Stem Cell. Jun 2;29 (6):905-917
Garcia-Aguilar J, Hernandez de Anda E, Sirivongs P, et al., A pathologic complete response to preoperative chemoradiation is associated with lower local recurrence and improved survival in rectal cancer patients treated by mesorectal excision. Dis Colon Rectum. Mar 2003;46(3):298-304
Yao Y, Xu X, Yang L, Zhu J, et al., Patient-Derived Organoids Predict Chemoradiation Responses of Locally Advanced Rectal Cancer. Cell Stem Cell. 2020;26(1):17-26 e6. Epub 2019/11/26
Ganesh K, Wu C, O'Rourke KP, Szeglin BC, et al., A rectal cancer organoid platform to study individual responses to chemoradiation. Nature Medicine. 2019;25(10):1607-14

Acknowledgements
Gabrielle Rupprecht, Hsu Lab members

Funding Sources
Discretionary funds

Affiliations
Duke Cancer Institute